Survival Update from a Multicenter, Randomized, Phase 3 Trial of Vitespen vs. Observation as Adjuvant Therapy For Renal Cell Carcinoma in Patients at High Risk Of Recurrence

Protocol C-100-27: International Survival Follow-up of Patients Enrolled In the Renal Cell Carcinoma Evaluation of Adjuvant Vitespen Treatment in the Antigenics C-100-12 (Part 1) (INSPIRE)

Christopher Wood,[1] Pramod Srivastava,[2] Louis Lacombe,[3] Andrei I Gorelov,[4] Sergei Gorelov,[5] Peter Mulders,[6] Henryk Zielinski,[7] Florentina Teofilovici,[8] Leah Isakov,[8] Bernard Escudier[9]

[1]M. D. Anderson Cancer Center, Houston, TX, USA; [2]University of Connecticut School of Medicine, Farmington, CT, USA; [3]CHUQ-Hôtel-Dieu de Québec, Pavillon de Recherche de L'Hôpital I'Hôtel Dieu, Quebec City, Quebec, Canada; [4]St. Petersburg State University, St. Petersburg, Russia; [5]St. Petersburg Hospital 122, St. Petersburg, Russia; [6]UMC St. Radboud, Nijmegen, Netherlands; [7]Centralny Szpital Kliniczny WAM, Warsaw, Poland; [8]Antigenics, Lexington, MA, USA; [9]Institut Gustave Roussy, Villejuif, France

INSPIRE (C-100-27) Protocol Design and Objectives

- **Background and Rationale**
 - Favorable recurrence-free survival (RFS) findings in earlier-stage patients enrolled in the Phase 3 study of vitespen in localized Renal Cell Carcinoma (Wood *et al.*, 2008) provided compelling evidence to continue follow-up for the secondary endpoint of overall survival. This is the largest study completed to date in the adjuvant setting.
- **Primary Objective**
 - To evaluate overall survival of patients previously enrolled in the C-100-12 protocol: A Multicenter, Randomized Phase 3 Study of Adjuvant Vitespen Versus Observation in Patients With High Risk of Recurrence After Surgical Treatment for Renal Cell Carcinoma
- **Study Period**
 - 3 years from last analysis point of 30 March 2007 (closure date of C-100-12 protocol)
 - Provides 9 years of total follow-up from time 1st patient randomized in C-100-12
- **Methodology**
 - Patient contact every 6 months if no disease recurrence, and every 3 months if disease has recurred

1 STUDY DESIGN AND PATIENTS

Study Design: From C-100-12 to C-100-27 Registry

C-100-12
Randomized Phase 3 Study of Adjuvant Vitespen vs Observation in Subjects With High Risk of Recurrence After Surgical Treatment for Renal Cell Carcinoma

C-100-27
International Survival Follow-up of Patients Enrolled In the RCC Evaluation of Adjuvant Vitespen Treatment in C-100-12 Protocol. **(INSPIRE)**

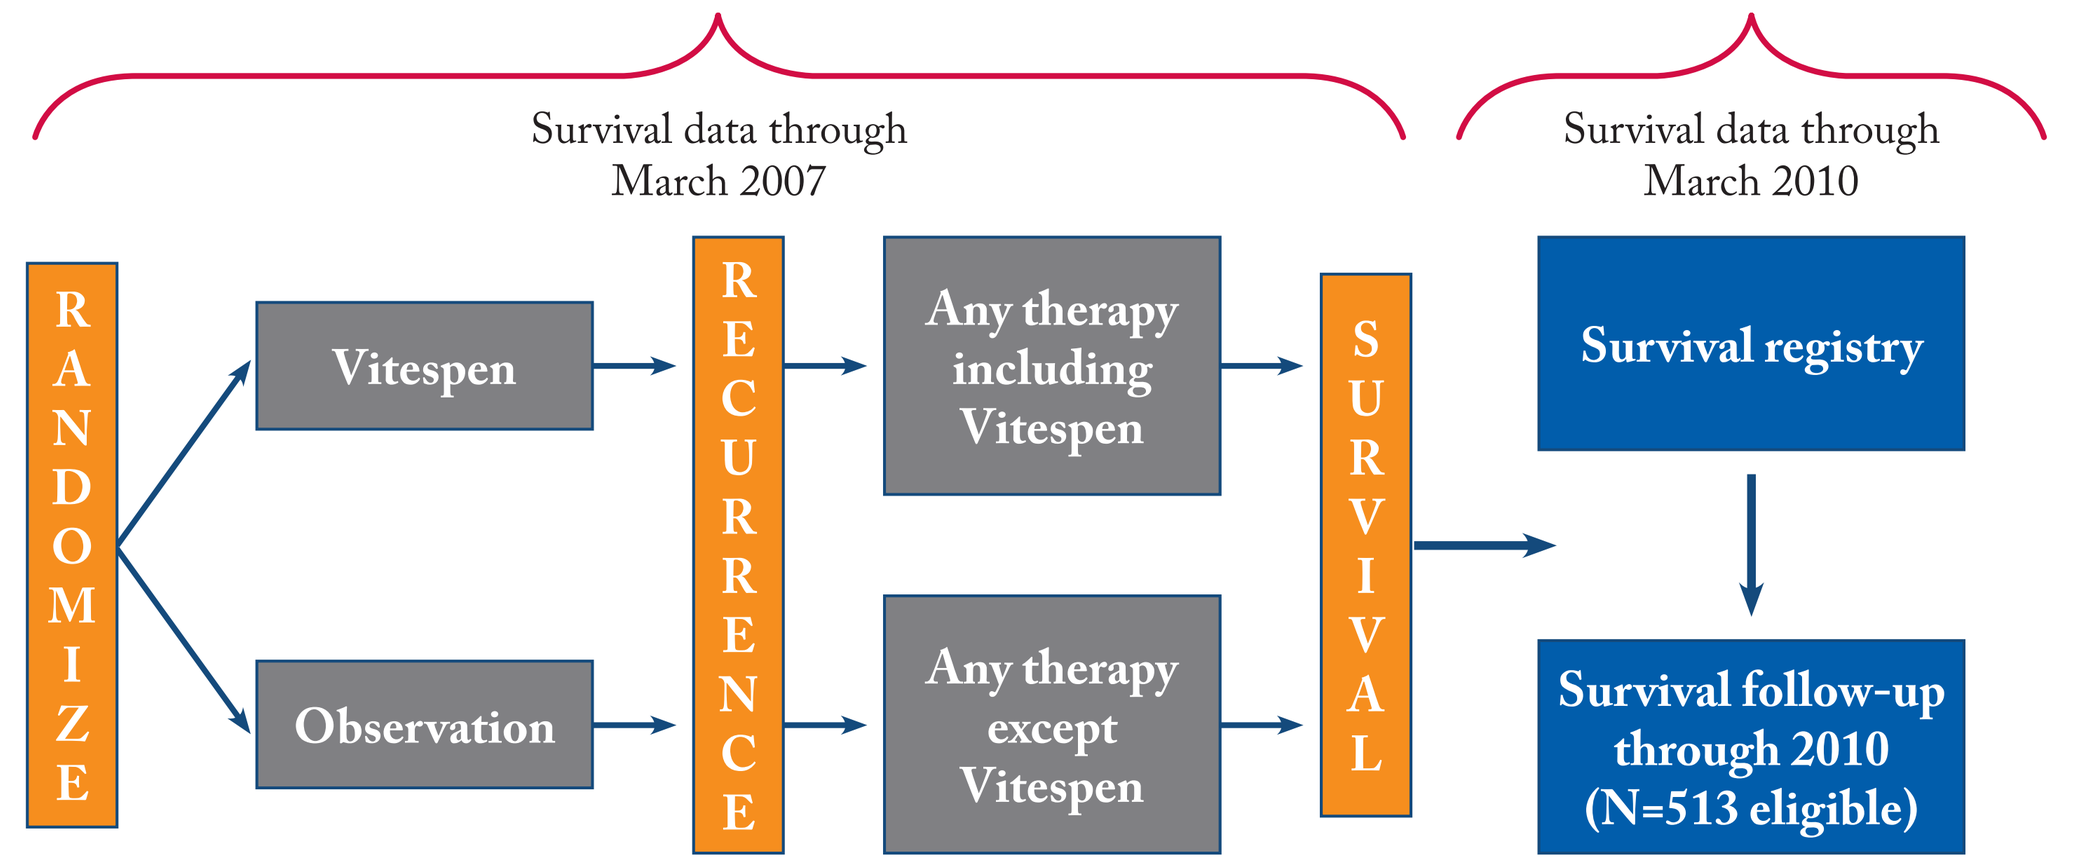

Survival data through March 2007
Survival data through March 2010
RANDOMIZE
Vitespen
Observation
RECURRENCE
Any therapy including Vitespen
Any therapy except Vitespen
SURVIVAL
Survival registry
Survival follow-up through 2010 (N=513 eligible)

Enrollment Update

- 513 of 728 randomized patients from C-100-12 potentially eligible for enrollment/contact
 - Attrition expected due to length of study, administrative barriers, lack of physician/patient cooperation
- Ongoing accrual as of 4 May 2009:
 - 56 clinical sites are active which represents approximately 429 potential patients (83% of eligible patient population and)
 - 341 (66.5%)of the eligible patients have been contacted
 - Active contact is ongoing

Statistical Methods

- Survival time is defined as the time from randomization into the clinical study C-100-12 protocol until death from any cause
- There are 513 eligible patients
- Final analysis: end of 3-year follow-up period
- Interim analysis: at 1.5 years (05 January 2009)
- Descriptive information is continually updated
- Overall survival times will be estimated using Kaplan-Meier methods
- Reported data will include:
 - Model based quartile estimates of survival time, HR, 95% CI and p-value based on log-rank test

Table 1. Patient Status in Study C-100-12 and Eligible Population for C-100-27

	Vitespen	Observation	Total
Total number of patients	361 (100%)	367 (100%)	728 (100%)
Death	70 (19.4%)	72 (19.6%)	142 (19.5%)
Administrative closure	256 (70.9%)	257 (70.0%)	513 (70.5%)
WOC	22 (6.1%)	19 (5.2%)	41 (5.6%)
LTF	13 (3.6%)	19 (5.2%)	32 (4.4%)
Registry Eligible	256 (100%)	257 (100%)	513 (100%)
Available data as of 5 Jan 2009	153 (59.8%)	153 (59.5%)	306 (59.6%)
FAS	138 (53.9%)	138 (53.7%)	276 (53.8%)
Intermediate Risk	95 (37.1%)	89 (34.6%)	184 (35.9%)

2 RESULTS

Overall Survival: Intent-to-Treat

Subgroup	Vitespen	Observation	HR (95% CI)	P Value
	N	N		
ITT	361	367		
Events Mar. 07	70 (19.4%)	72 (19.6%)	0.978 (0.702, 1.364)	0.896
Events Jan. 09	83 (23.0%)	90 (24.5%)	0.940 (0.696, 1.271)	0.688
Median Follow-up time	4.2 years	4.1 years		

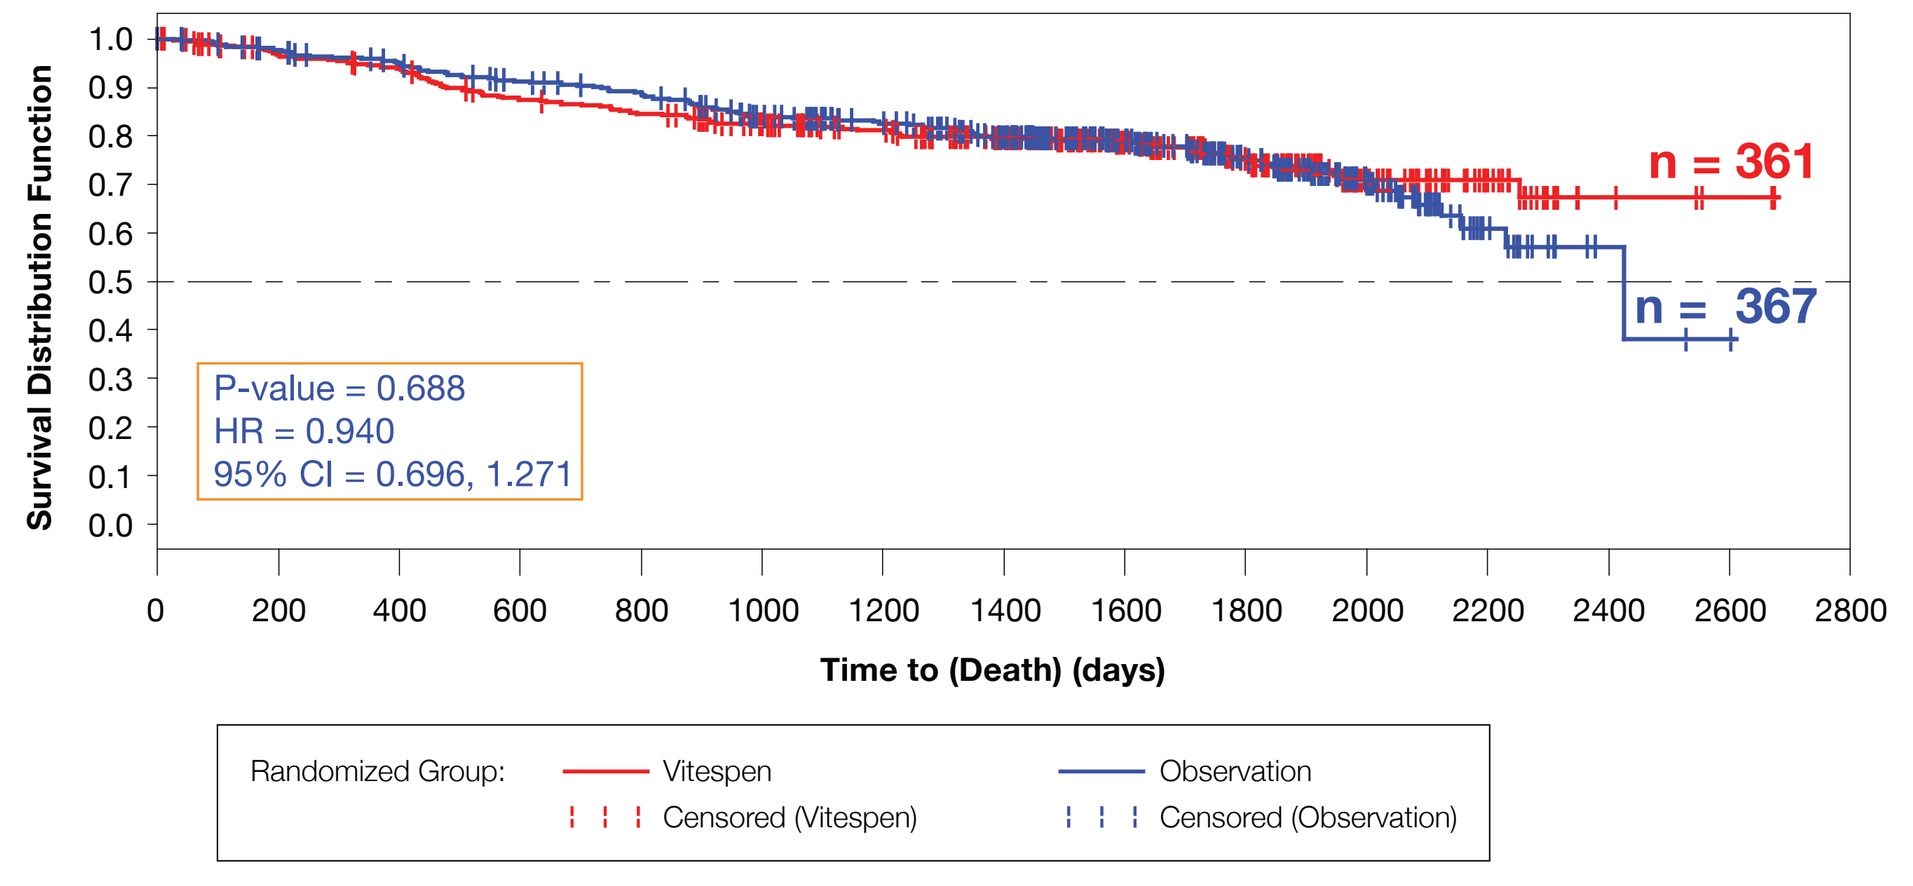

Survival Distribution Function
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
n = 361
n = 367
P-value = 0.688
HR = 0.940
95% CI = 0.696, 1.271
0 200 400 600 800 1000 1200 1400 1600 1800 2000 2200 2400 2600 2800
Time to (Death) (days)
Randomized Group:
Vitespen
Censored (Vitespen)
Observation
Censored (Observation)

Overall Survival: Full Analysis Set

Subgroup	Vitespen	Observation	HR (95% CI)	P Value
	N	N		
FAS	300	304		
Events Mar. 07	37 (12.3%)	40 (13.2%)	0.822 (0.561, 1.388)	0.586
Events Jan. 09	44 (14.7%)	51 (16.8%)	0.813 (0.538, 1.229)	0.326
Median Follow-up time	4.4 years	4.3 years		

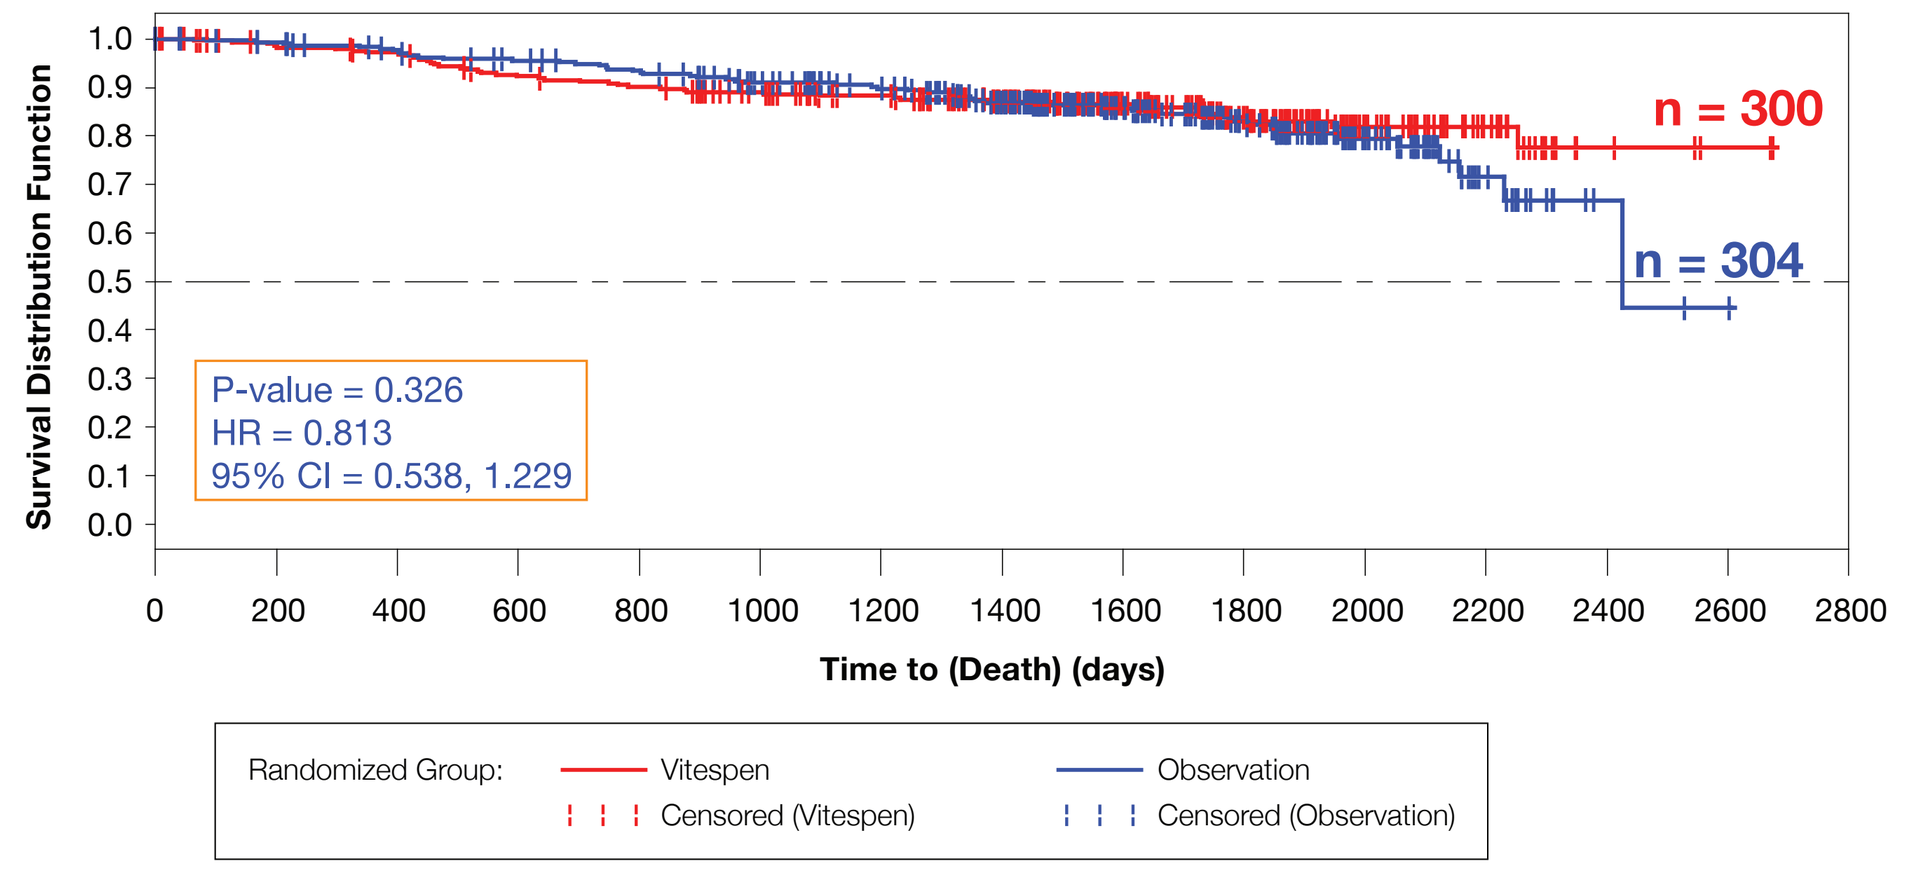

Survival Distribution Function
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
n = 300
n = 304
P-value = 0.326
HR = 0.813
95% CI = 0.538, 1.229
0 200 400 600 800 1000 1200 1400 1600 1800 2000 2200 2400 2600 2800
Time to (Death) (days)
Randomized Group:
Vitespen
Censored (Vitespen)
Observation
Censored (Observation)

Overall Survival: AJCC Stage I+II
Earlier Stages of Disease

Subgroup	Vitespen	Observation	HR (95% CI)	P Value
	N	N		
AJCC Stage I+II	125	115		
Events Mar. 07	12 (9.6%)	16 (13.9%)	0.717 (0.339, 1.517)	0.382
Events Jan. 09	13 (10.4%)	20 (17.4%)	0.628 (0.312, 1.266)	0.190
Median Follow-up time	4.4 years	4.2 years		

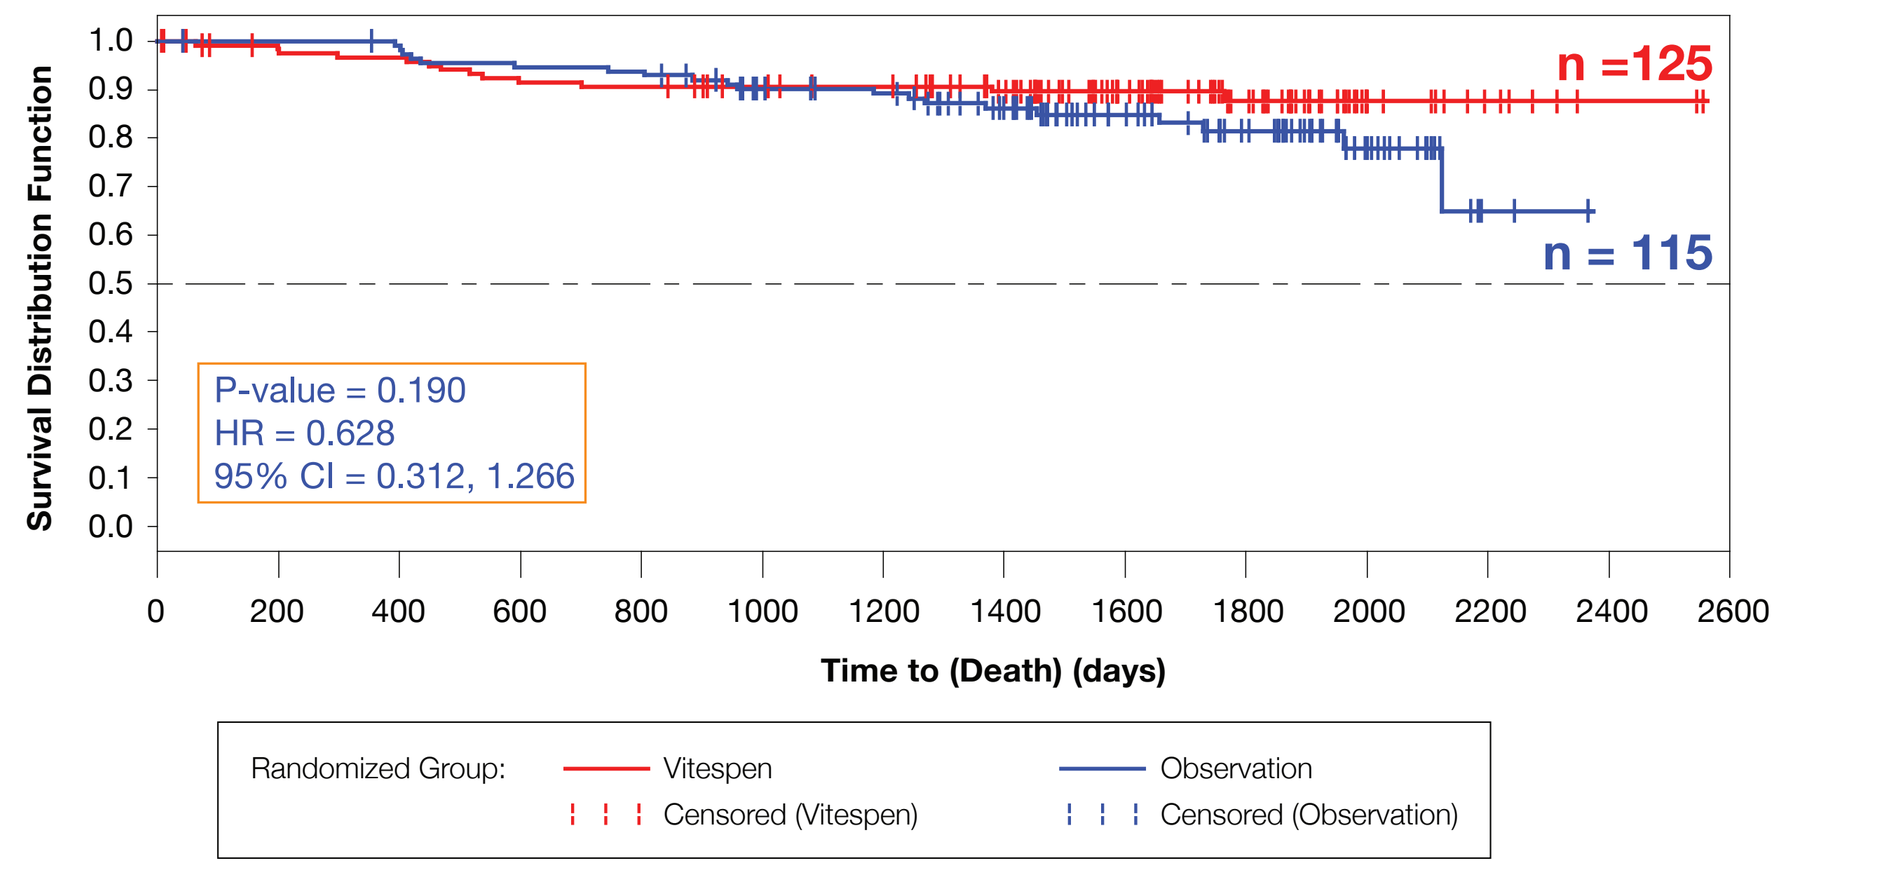

Survival Distribution Function
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
n =125
n = 115
P-value = 0.190
HR = 0.628
95% CI = 0.312, 1.266
0 200 400 600 800 1000 1200 1400 1600 1800 2000 2200 2400 2600
Time to (Death) (days)
Randomized Group:
Vitespen
Censored (Vitespen)
Observation
Censored (Observation)

Overall Survival: Intermediate Risk*
Earlier Stages of Disease

Subgroup	Vitespen	Observation	HR (95% CI)	P Value
	N	N		
Intermediate Risk	184	178		
Events Mar. 07	15 (8.2%)	25 (14.0%)	0.608 (0.320, 1.156)	0.126
Events Jan. 09	**18 (9.8%)**	**32 (18.0%)**	**0.541 (0.303, 0.967)**	**0.036**
Median Follow-up time	4.5 years	4.3 years		

*AJCC Stage I (T1b), AJCC Stage II (T2) high-grade, Stage III (T3a/T1/T2) low grade

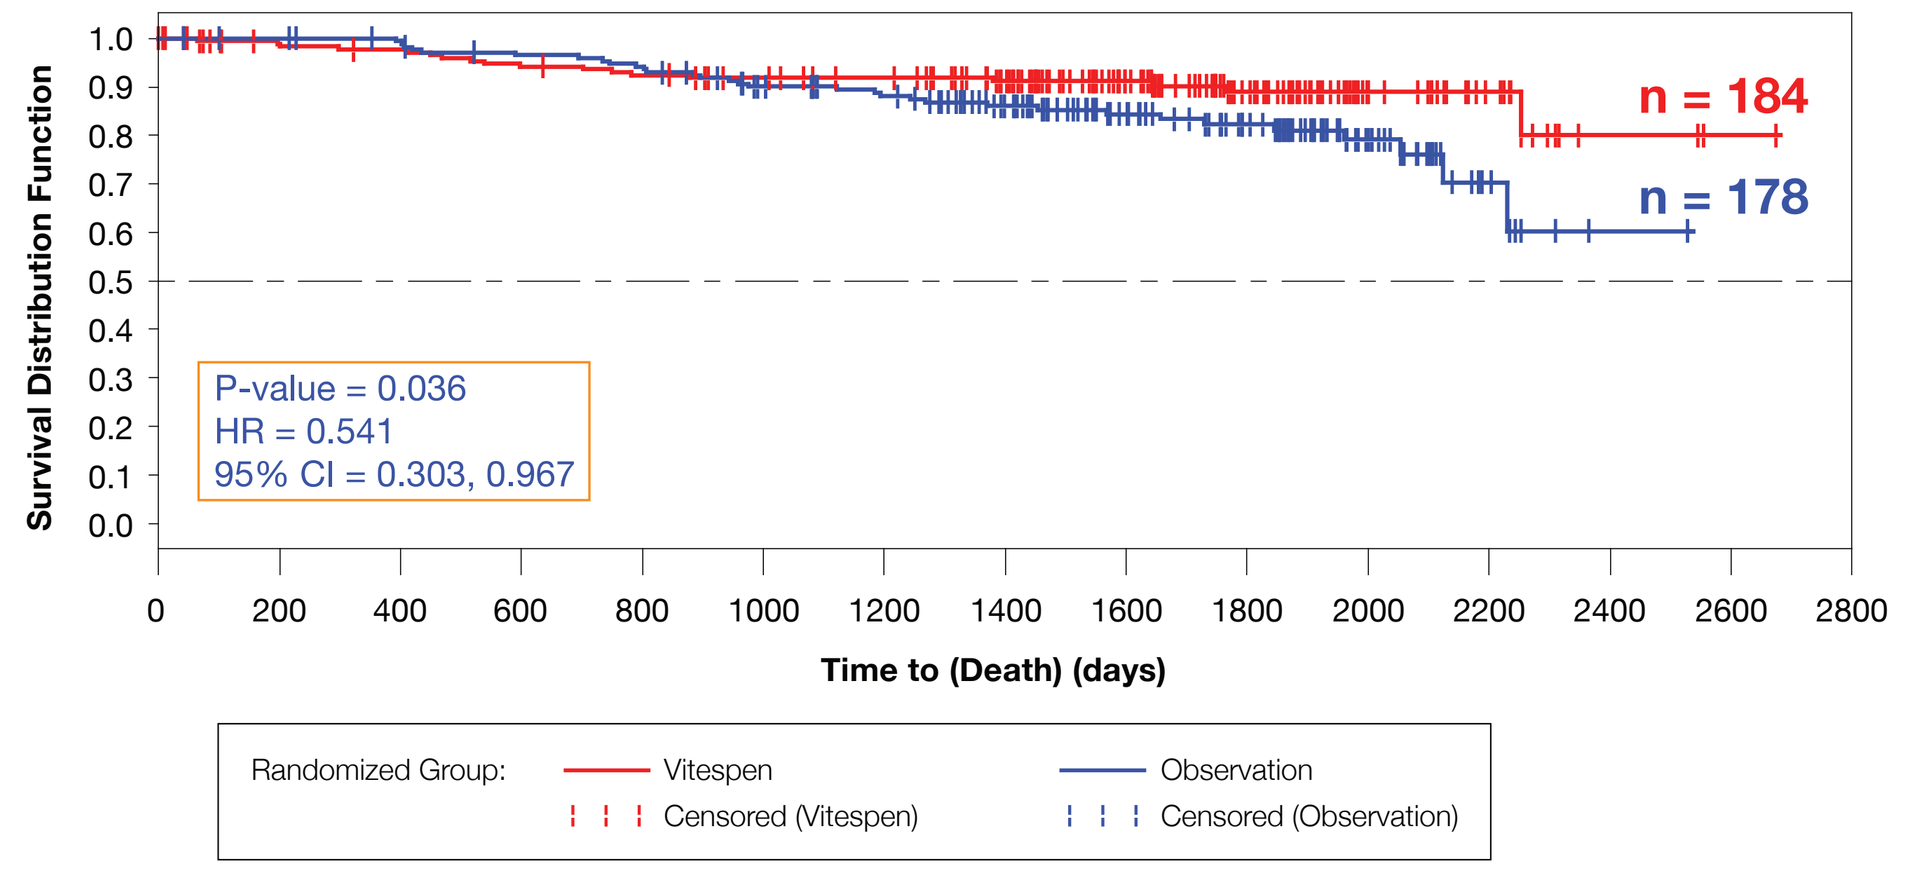

Survival Distribution Function
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
n = 184
n = 178
P-value = 0.036
HR = 0.541
95% CI = 0.303, 0.967
0 200 400 600 800 1000 1200 1400 1600 1800 2000 2200 2400 2600 2800
Time to (Death) (days)
Randomized Group:
Vitespen
Censored (Vitespen)
Observation
Censored (Observation)

Table 2. Disposition by Prior C-100-12 Population and Baseline Disease Status as of 5 January 2009

	Vitespen	Observation	Total
Patients Eligible for Registry (ITT)[a]	256 (100.0%)	257 (100.0%)	513 (100.0%)
In the Database[b] (Data Transfer 05 Jan 2009)	153 (59.8%)	153 (59.5%)	306 (59.6%)
Investigator-Reported Recurrence	35 (13.7%)	46 (17.9%)	81 (15.8%)
Baseline Disease[c]	15 (5.9%)	15 (5.8%)	30 (5.8%)
Full Analysis Set[d]	138 (53.9%)	138 (53.7%)	276 (53.8%)
AJCC Stage I/II	60 (23.4%)	58 (22.6%)	118 (23.0%)
AJCC Stage I	2 (0.8%)	2 (0.8%)	4 (0.8%)
AJCC Stage II	58 (22.7%)	56 (21.8%)	114 (22.2%)
AJCC Stage III	72 (28.1%)	75 (29.2%)	147 (28.7%)
AJCC Stage IV	6 (2.3%)	5 (1.9%)	11 (2.1%)
Intermediate-Risk[e]	95 (37.1%)	89 (34.6%)	184 (35.9%)

ITT: intent-to-treat; AJCC: American Joint Committee on Cancer.
a Includes randomized eligible patients who discontinued the study due to administrative study closure.
b There are 12 patients in the database (8 vitespen, 4 observation) with missing data or were lost to follow-up. Also, 2 patients who were recorded as lost to follow-up in the C-100-12 study were located and are participating in the C-100-27 protocol.
c As determined by CEC review.
d The full analysis set (n = 604) excludes the 124 patients who were deemed by the CEC to have residual or metastatic disease at baseline.
e Intermediate-risk is defined as patients with AJCC stage I or stage II (T1/T2) disease with high Fuhrman grade or AJCC stage III T1/T2/T3a disease with low Fuhrman grade.

Table 3. Baseline Demographic Characteristics of C-100-27 Patients

	Vitespen (n = 145)	Observation (n = 149)	Total (N = 294)*
Age			
Mean (SD)	58.6 (11.0)	59.3 (9.95)	59.0 (10.5)
Median	58.0	59.0	59.0
Min, Max	29, 81	36, 83	29, 83
Gender [n(%)]			
Male	85 (58.6)	86 (57.7)	171 (58.2)
Female	60(41.4)	63 (42.3)	123 (41.8)
Race [n(%)]			
Caucasian/White	139 (95.9)	146 (98.0)	285 (96.9)
Hispanic	3 (2.1)	1 (0.7)	4 (1.4)
Asian/Pacific	1 (0.7)	1 (0.7)	2 (0.7)
Black	–	1 (0.7)	1 (0.3)
Other	2 (1.4)	–	2 (0.7)
Performance Status [n(%)]			
0	109 (75.2)	113 (75.8)	222 (75.5)
1	36 (24.8)	36 (24.2)	72 (24.5)

SD: standard deviation; min, max: minimum, maximum.
Note: Based on information collected at baseline in the C-100-12 protocol.
* There are 12 patients in the database with missing data or were lost to follow-up.

Table 4. Baseline Disease Characteristics of C-100-27 Patient as of 5 January 2009

		Vitespen (n = 145)	Observation (n = 149)	Total (N = 294)[a]
AJCC Stage [N(%)]	I	2 (1.4)	3 (2.0)	5 (1.7)
	II	58 (40.0)	60 (40.3)	118 (40.1)
	III	77 (53.1)	78 (52.3)	155 (52.7)
	IV	8 (5.5)	8 (5.4)	16 (5.4)
Tumor Stage[b] [N(%)]	T1	4 (2.8)	5 (3.4)	9 (3.1)
	T2	61 (42.1)	62 (41.6)	123 (41.8)
	T3a	46 (31.7)	46 (30.9)	92 (31.3)
	T3b	28 (19.3)	31 (20.8)	59 (20.1)
	T3c	5 (3.4)	2 (1.3)	7 (2.4)
	T4	1 (0.7)	3 (2.0)	4 (1.4)
Lymph Node Status[b] [N(%)]	NX or N0	133 (91.7)	139 (93.3)	272 (92.5)
	N+	12 (8.3)	10 (6.7)	22 (7.5)
Metastatic/Residual Disease at Baseline[c] [N(%)]	Yes	13 (9.0)	15 (10.1)	28 (9.5)
	No	132 (91.0)	134 (89.9)	266(90.5)
Nuclear Grade [N(%)]	Low (1–2)	54 (37.2)	45 (30.2)	99(33.7)
	High (3-4)	91 (62.8)	104 (69.8)	195 (66.3)

AJCC: American Joint Committee on Cancer.
Note: Based on information collected at baseline in the C-100-12 protocol.
All eligible patients had clear cell carcinoma.
a There are 12 patients in the database with missing data or were lost to follow-up.
b Determined for the purpose of analysis as the maximum of the pathological and clinical assessments.
c As determined by CEC review.

3 CONCLUSION

Conclusions

- Overall survival curves show consistent and similar trends to recurrence-free survival from Study C-100-12
- Overall survival trends support recurrence-free survival findings in earlier-stage patient groups
- Ongoing results support benefit of vitespen in overall survival for earlier-stage patient
- These results in earlier-stage patient groups are consistent with the biological rationale for cancer vaccine efficacy
- Patient accrual and follow-up time is equivalent between arms in all analysis sets